SDU Research Project

SDU

Advanced Raman Spectroscopy for Future Manufacturing of Biopharmaceuticals

PhD Student: Jeppe Hagedorn, SDU Biotechnology & Novo Nordisk A/S
Principal Supervisor: Martin A.B. Hedegaard, SDU Biotechnology
Company Supervisor: Lars Poulsen, Novo Nordisk A/S

The aim of this project is to further develop and implement Raman systems to enable production of better biopharmaceuticals. Achieving optimized Raman signals by Transmission Raman Spectroscopy in bioprocesses will act as the foundation for furthering the development of manufacturing of biopharmaceuticals.

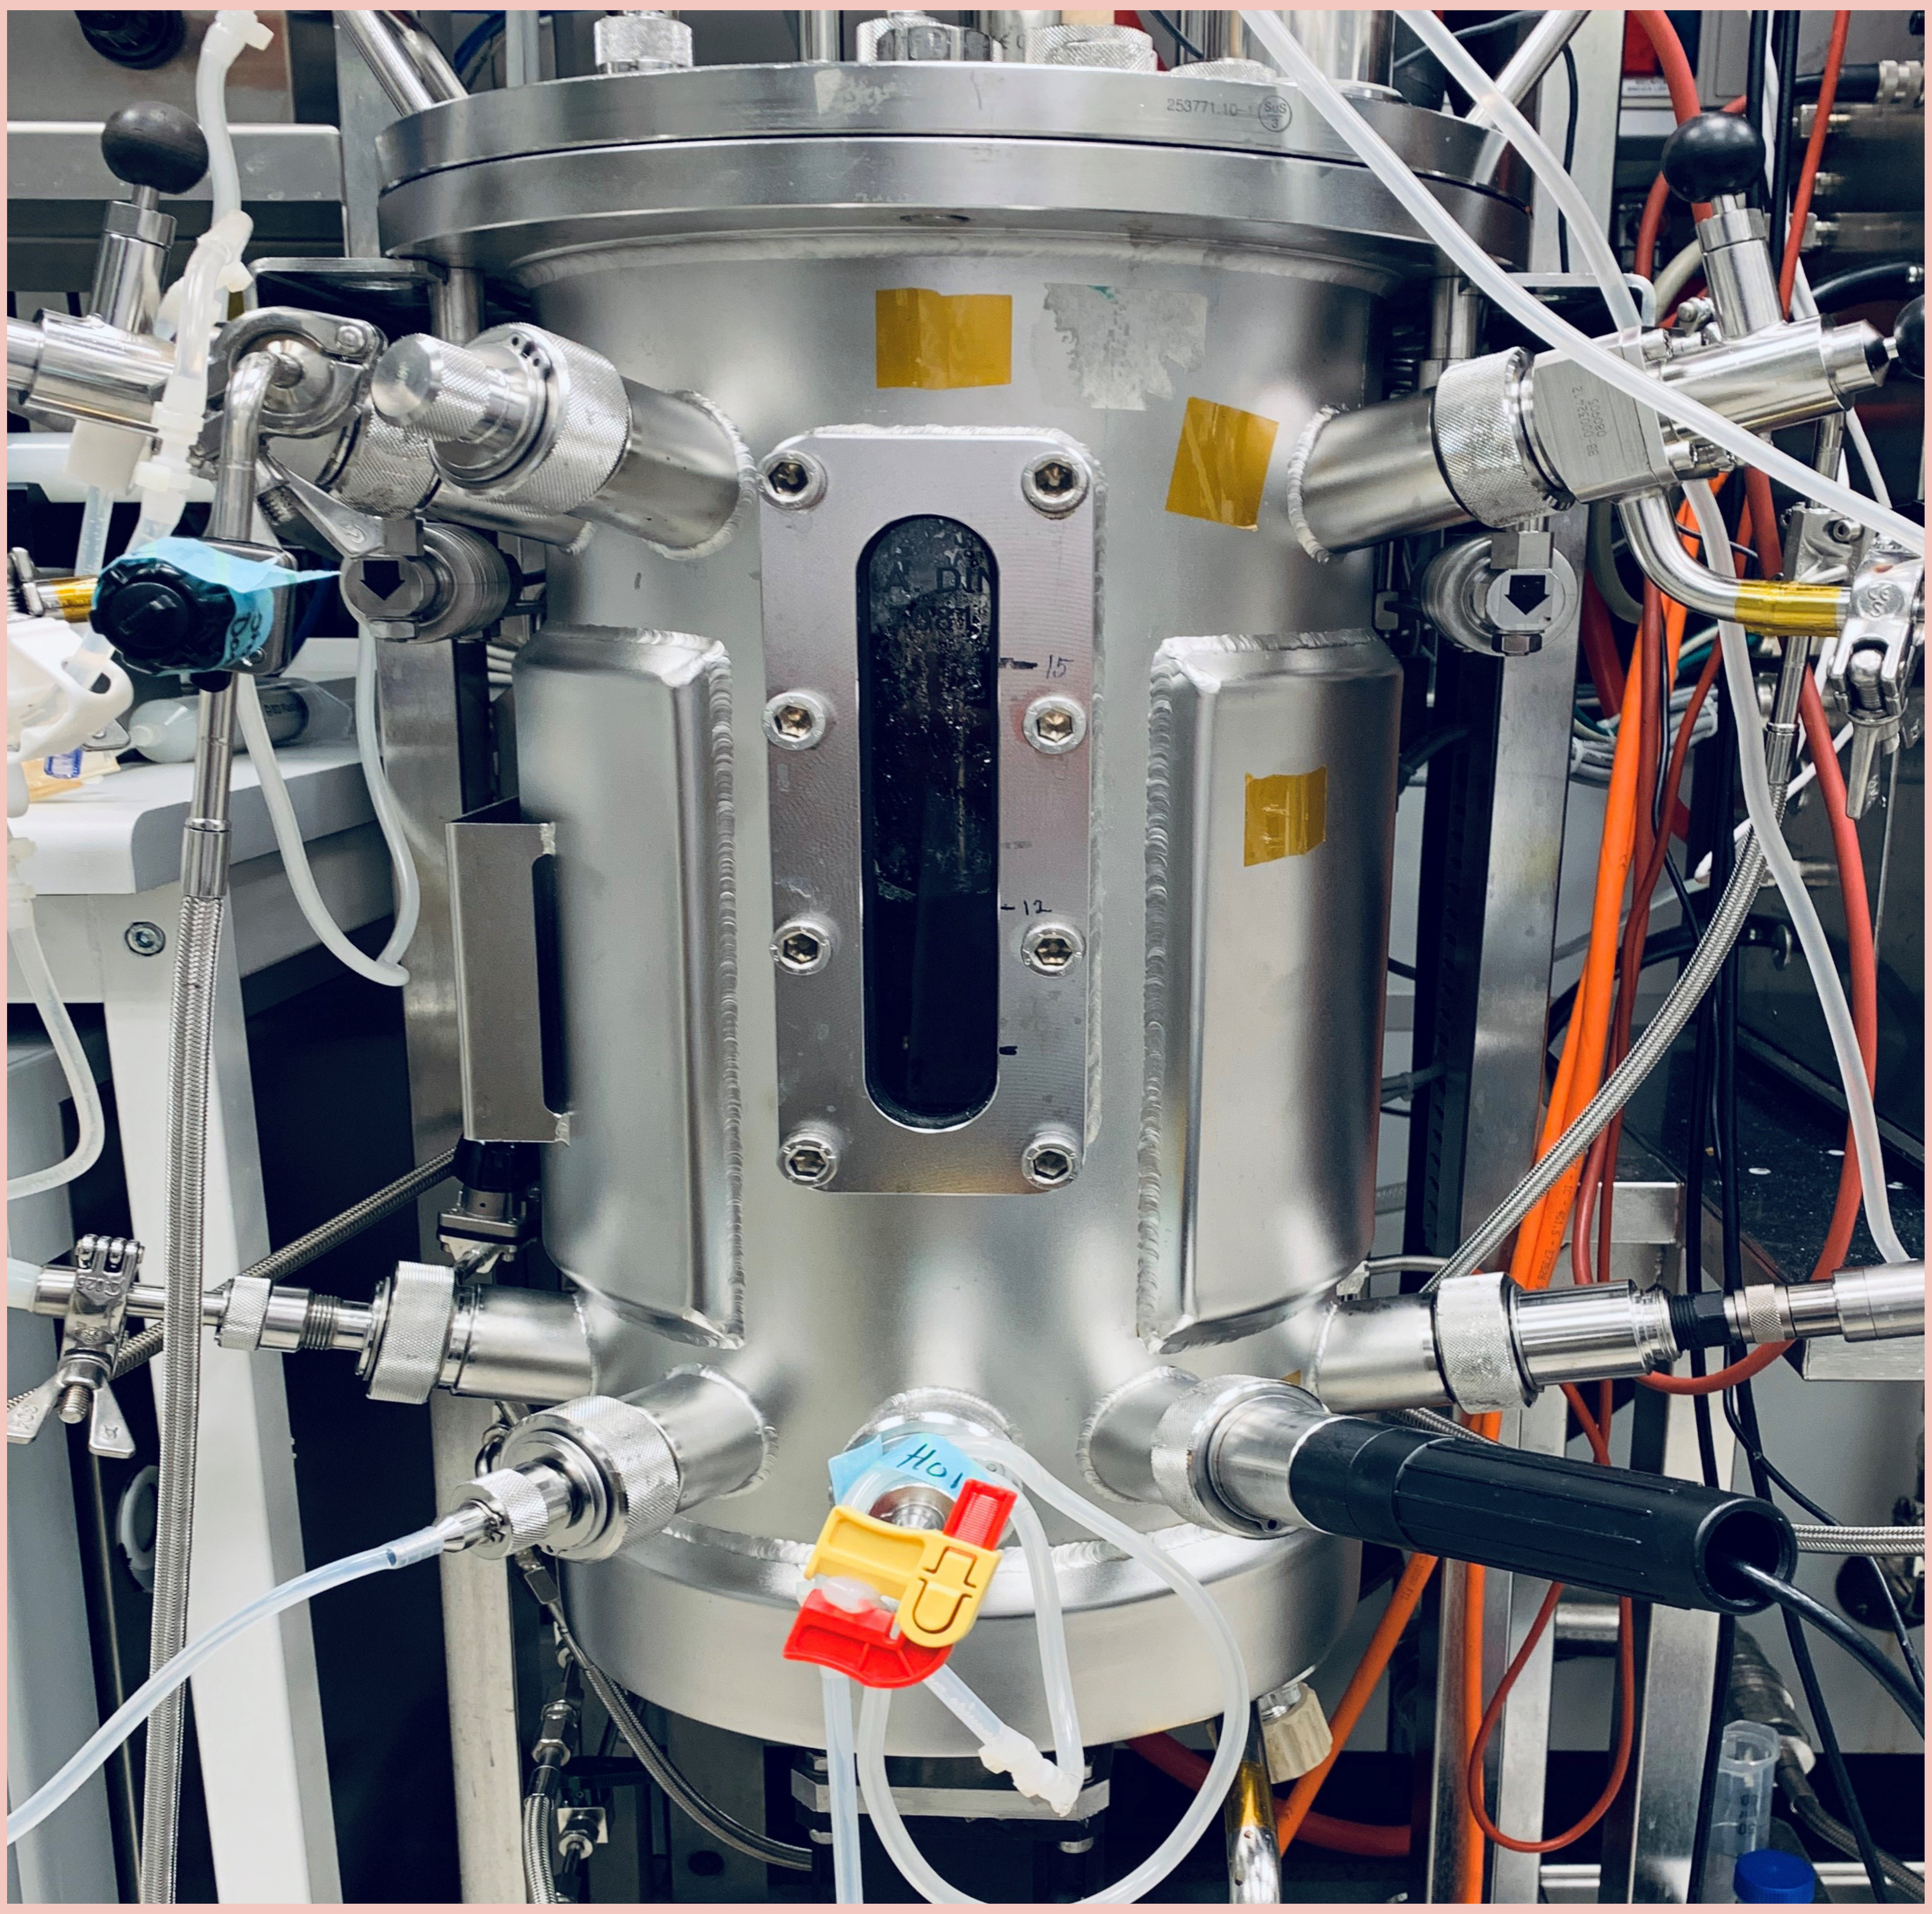
15
12
HO1

A 15 L bioreactor for production of monoclonal antibodies from mammalian cells for therapeutic use. © J. Hagedorn, 2020

1. Background

Raman spectroscopy as a quantitative analytical technique poses a significant upgrade to the current off-line chemical analysis [1], which require sample preparation known to introduce variation in the measurements, is time consuming, expensive, and destructive. The signal intensity is considerably lower for Raman compared with mid-IR spectroscopy and Raman does not have the benefit of being described by Beer-Lamberts law [2]. Hence, a significant number of reference samples are usually needed to establish accurate calibration models, and even that may not lead to optimal model development [3].

Achieving a significant increase in the Raman signals which are relevant for the specific applications within biopharma while decreasing the complexity of the modelling processes will help the scientific and industrial field significantly [4].

2. Objectives

With this project we aim for replacing current state-of-the-art analysis methods for chemical control of manufacturing processes with advanced Raman spectroscopy, by answering the following questions:

I. How to optimize Raman instrumentation, probe design and sampling interface to acquire the best possible signal from bioprocesses and replace the need for advanced data analysis methods?

The relevant sub-objectives and research questions are:

i. How can Transmission Raman spectroscopy aid in the determination of critical quality attributes for induced pluripotent stem cells?

ii. Can an optimized Raman signal via Transmission mode enable global models for biopharmaceutical processes? And what is the roadmap for the development of these models?

iii. How to best implement advanced control of mammalian cell cultures via Raman predictions?

iv. Can advanced machine learning concepts such as semi-supervised learning algorithms and non-linear methods improve accuracies and provide robust prediction models?

3. Methodology

A. Building a flexible Raman set-up, which will enable the comparisions needed to answer the research questions stated in Section 2. This instrument will be financed by Novo Nordisk A/S.

B. Statistical and multivariate data analysis/chemometrics, will form the basis of the data analysis performed throughout the project. It will be used in both an exploratory manner and for the development of calibration models which will be deployed in processes in Novo Nordisk A/S.

C. When required, cultivations in lab-scale (or larger) bioreactors, such as the one seen in the picture above will be performed.

D. For reference analysis chromatographic separation may be required and, if necessary, methods may be developed.

4. References

1. Raman, C.V., and K.S. Krishnan. 1928. "A new type of secondary radiation." Nature.
2. Diem, Max. 2015. Modern Vibrational Spectroscopy and Micro-Spectroscopy: Theory, Instrumentation and Biomedical Applications. Chichester, UK: John Wiley & Sons, Ltd.
3. André, Silvère, Sylvian Lagresle, Zahia Hannas, Éric Calvosa, and Ludovic Duponchel. 2017. "Mammalian cell culture monitoring using in-situ spectroscopy: Is your method really optimised?" Biotechnology Progress 308-316.
4. Buckley, Kevin, and Alan G. Ryder. 2017. "Applications of Raman spectroscopy in Biopharmaceutical Manufacturing: A short Review." Applied Spectroscopy 1085-1116.

sdu.dk

Hagedorn, J., University of Southern Denmark

novo nordisk®